NKX101, an allogeneic off-the-shelf CAR NK cell therapy targeting NKG2D-Ls, has potent anti-leukemic activity alone or in combination with Ara-C

Cynthia Cho, Nikki Kimura, Jessica Sood, Damie Juat, Max Zhang, Carmel Chan, Ph.D., Tao Geng Ph.D, Kyle Hansen, James Trager, Ph.D.

Abstract Presentation Number: 3603

Introduction

NKG2D ligands (NKG2D-L) are a family of immunomodulatory proteins that are upregulated on damaged or transformed cells and have limited expression on normal tissues. Natural killer (NK) cells target and eliminate NKG2D-L^{+} cells through NKG2D receptor binding and subsequent activation. NKX101 is an allogeneic off-the-shelf healthy donor-derived chimeric antigen receptor (CAR) NK cell therapy candidate engineered to express a NKG2D-L targeting CAR and membrane-bound IL-15 for extended persistence. In a clinical trial, NKX101 treatment combined with modified lymphodepletion incorporating cytarabine (Ara-C) has shown promise for the treatment of relapsed/refractory (r/r) AML, including generating complete responses (CR) with MRD negativity (MRD^{-}). To support the rationale of NKX101 combined with modified lymphodepletion, we developed assays to (i) investigate the cytotoxic activity of NKX101 against individual NKG2D-Ls, (ii) determine the effect of Ara-C pre-treatment on NKG2D-L expression on AML cell lines, (iii) evaluate the combined anti-leukemic activity of NKX101 and Ara-C, and (iv) assess NKX101 cytotoxicity of Ara-C resistant cell lines.

Engineered CHO-K1 cells express high levels of NKG2D-L and have similar growth kinetics compared to CHO-K1 empty vector controls

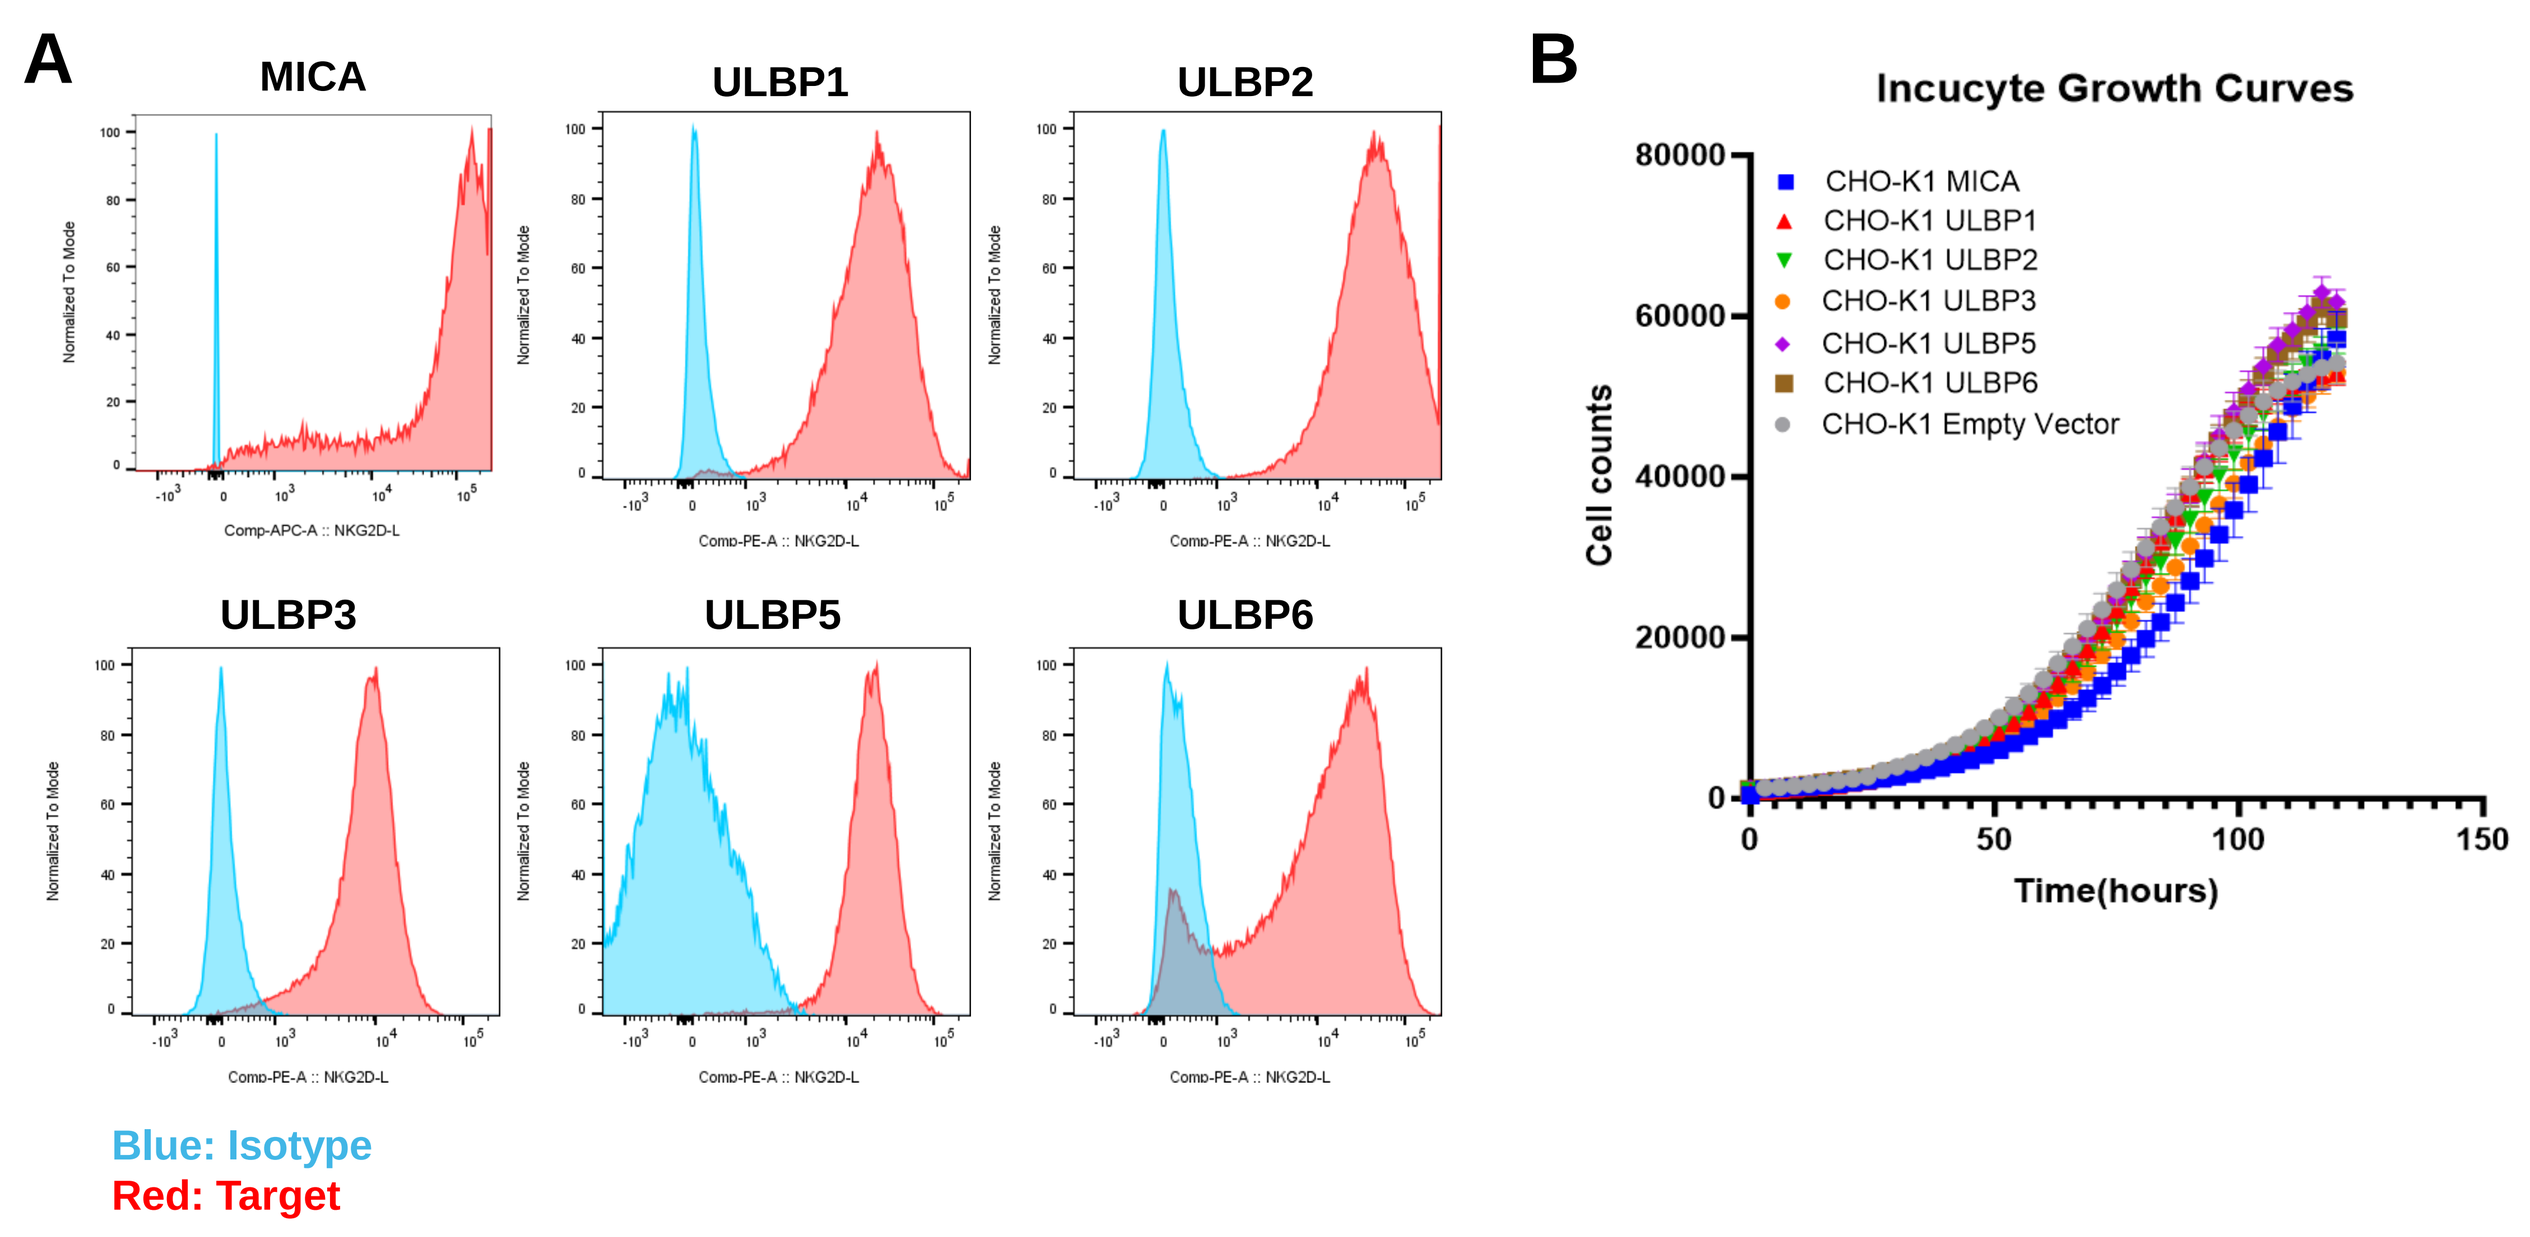
A
MICA
ULBP1
ULBP2
ULBP3
ULBP5
ULBP6
B
Incucyte Growth Curves
CHO-K1 MICA
CHO-K1 ULBP1
CHO-K1 ULBP2
CHO-K1 ULBP3
CHO-K1 ULBP5
CHO-K1 ULBP6
CHO-K1 Empty Vector
Cell counts
Time(hours)
Blue: Isotype
Red: Target

Figure 1. A) NKG2D ligand expression on engineered CHO-K1 cells. B) Growth curves of NucRed™ labeled engineered CHO-K1 cells expressing a single NKG2D ligand.

NKX101 cells potently target CHO-K1 cells engineered to express individual NKG2D-Ls

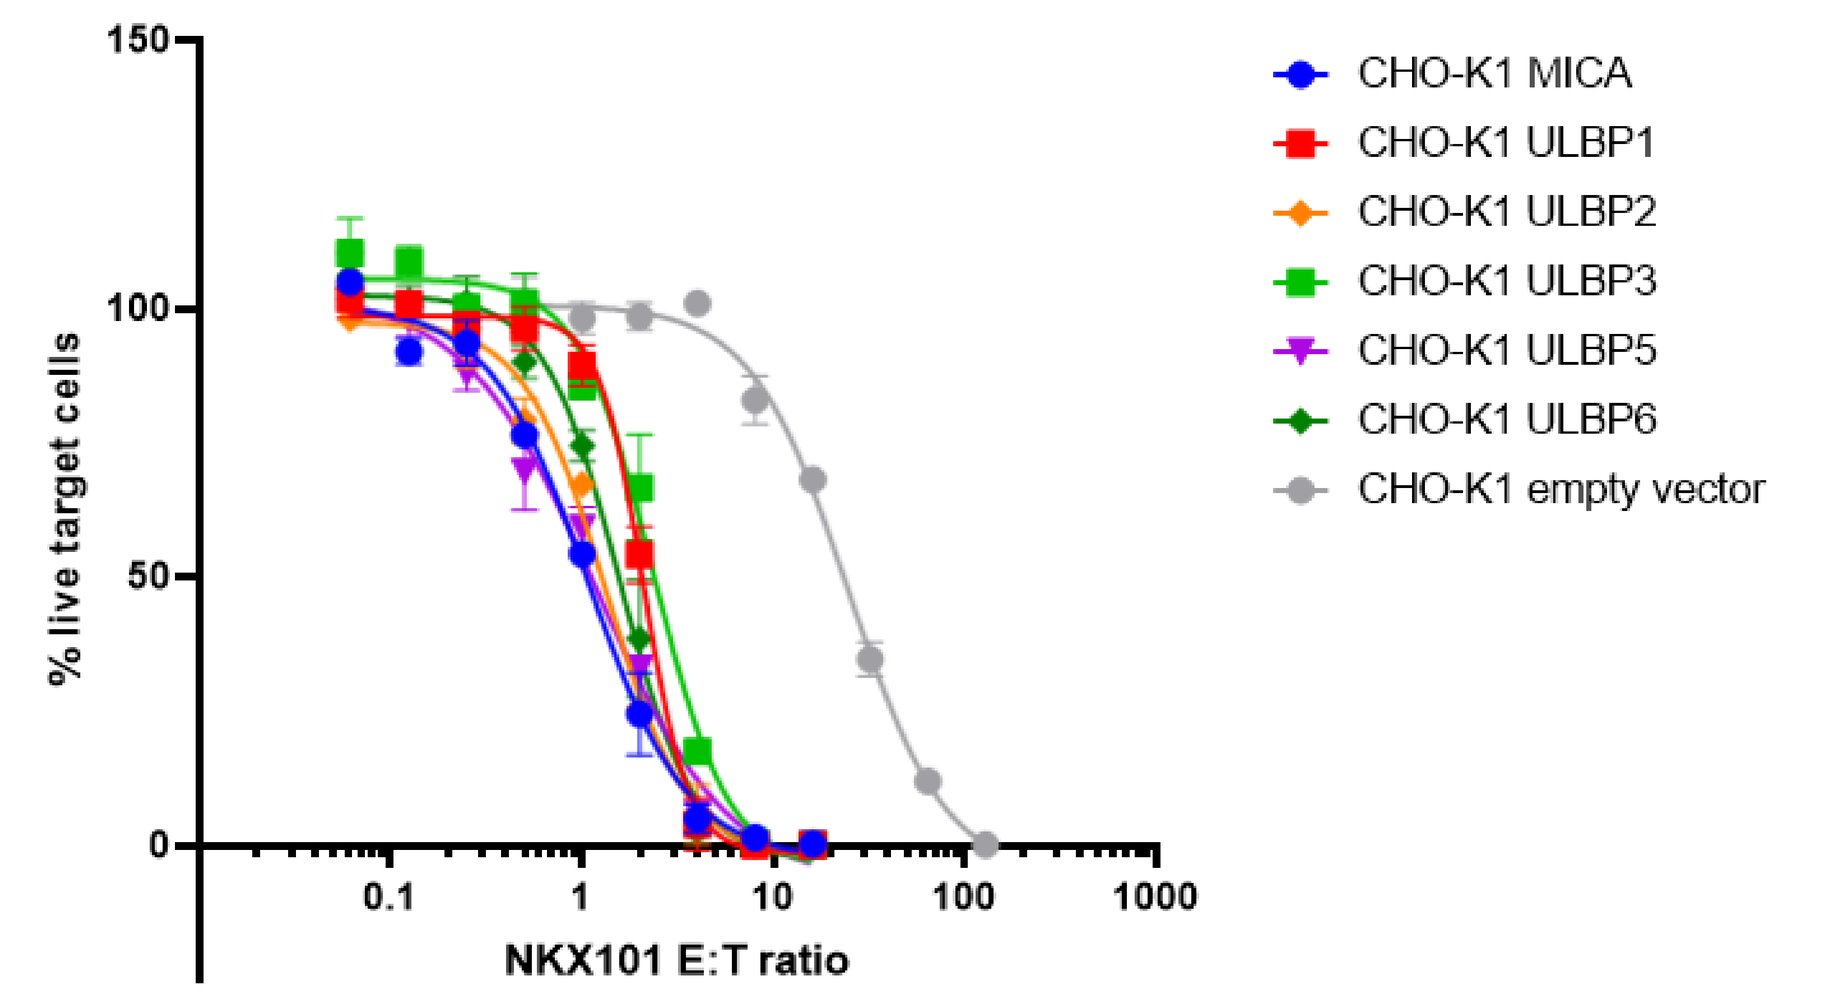
CHO-K1 MICA
CHO-K1 ULBP1
CHO-K1 ULBP2
CHO-K1 ULBP3
CHO-K1 ULBP5
CHO-K1 ULBP6
CHO-K1 empty vector
% live target cells
NKX101 E:T ratio

	CHO-K1 MICA	CHO-K1 ULBP1	CHO-K1 ULBP2	CHO-K1 ULBP3	CHO-K1 ULBP5	CHO-K1 ULBP6	CHO-K1 empty vector
IC50	1.07	2.09	1.33	2.36	1.189	1.56	24.5

Figure 2. Targeted killing of CHO-K1 cells expressing a single NKG2D-L or empty vector control.

Methods

NKX101 cells were generated from peripheral blood leukopaks from healthy donors. CHO-K1 cells were engineered to stably express individual NKG2D-Ls or empty vector controls and were co-cultured with NKX101 cells for cytotoxicity experiments. AML cell lines were treated with a dose titration of Ara-C and evaluated for NKG2D-L expression by flow cytometry. To measure NKX101 cytotoxicity against AML cell lines, NucRed™-labeled target cells were pre-treated with Ara-C using IC_{50} and IC_{90} concentrations and co-cultured with NKX101. Specific cell killing of target cells was measured using an Incucyte® instrument for five days. The cytotoxic effect of each agent was determined using a dose-response matrix of increasing concentrations of NKX101 cells and Ara-C to generate specific cytotoxicity values. Combination effects were analyzed using the SynergyFinder Plus web application. Ara-C resistant HL-60 and THP-1 cell lines were established by treating the cells with increasing concentrations of Ara-C over the span of several months.

Ara-C treatment upregulates NKG2D-L expression on AML cell lines

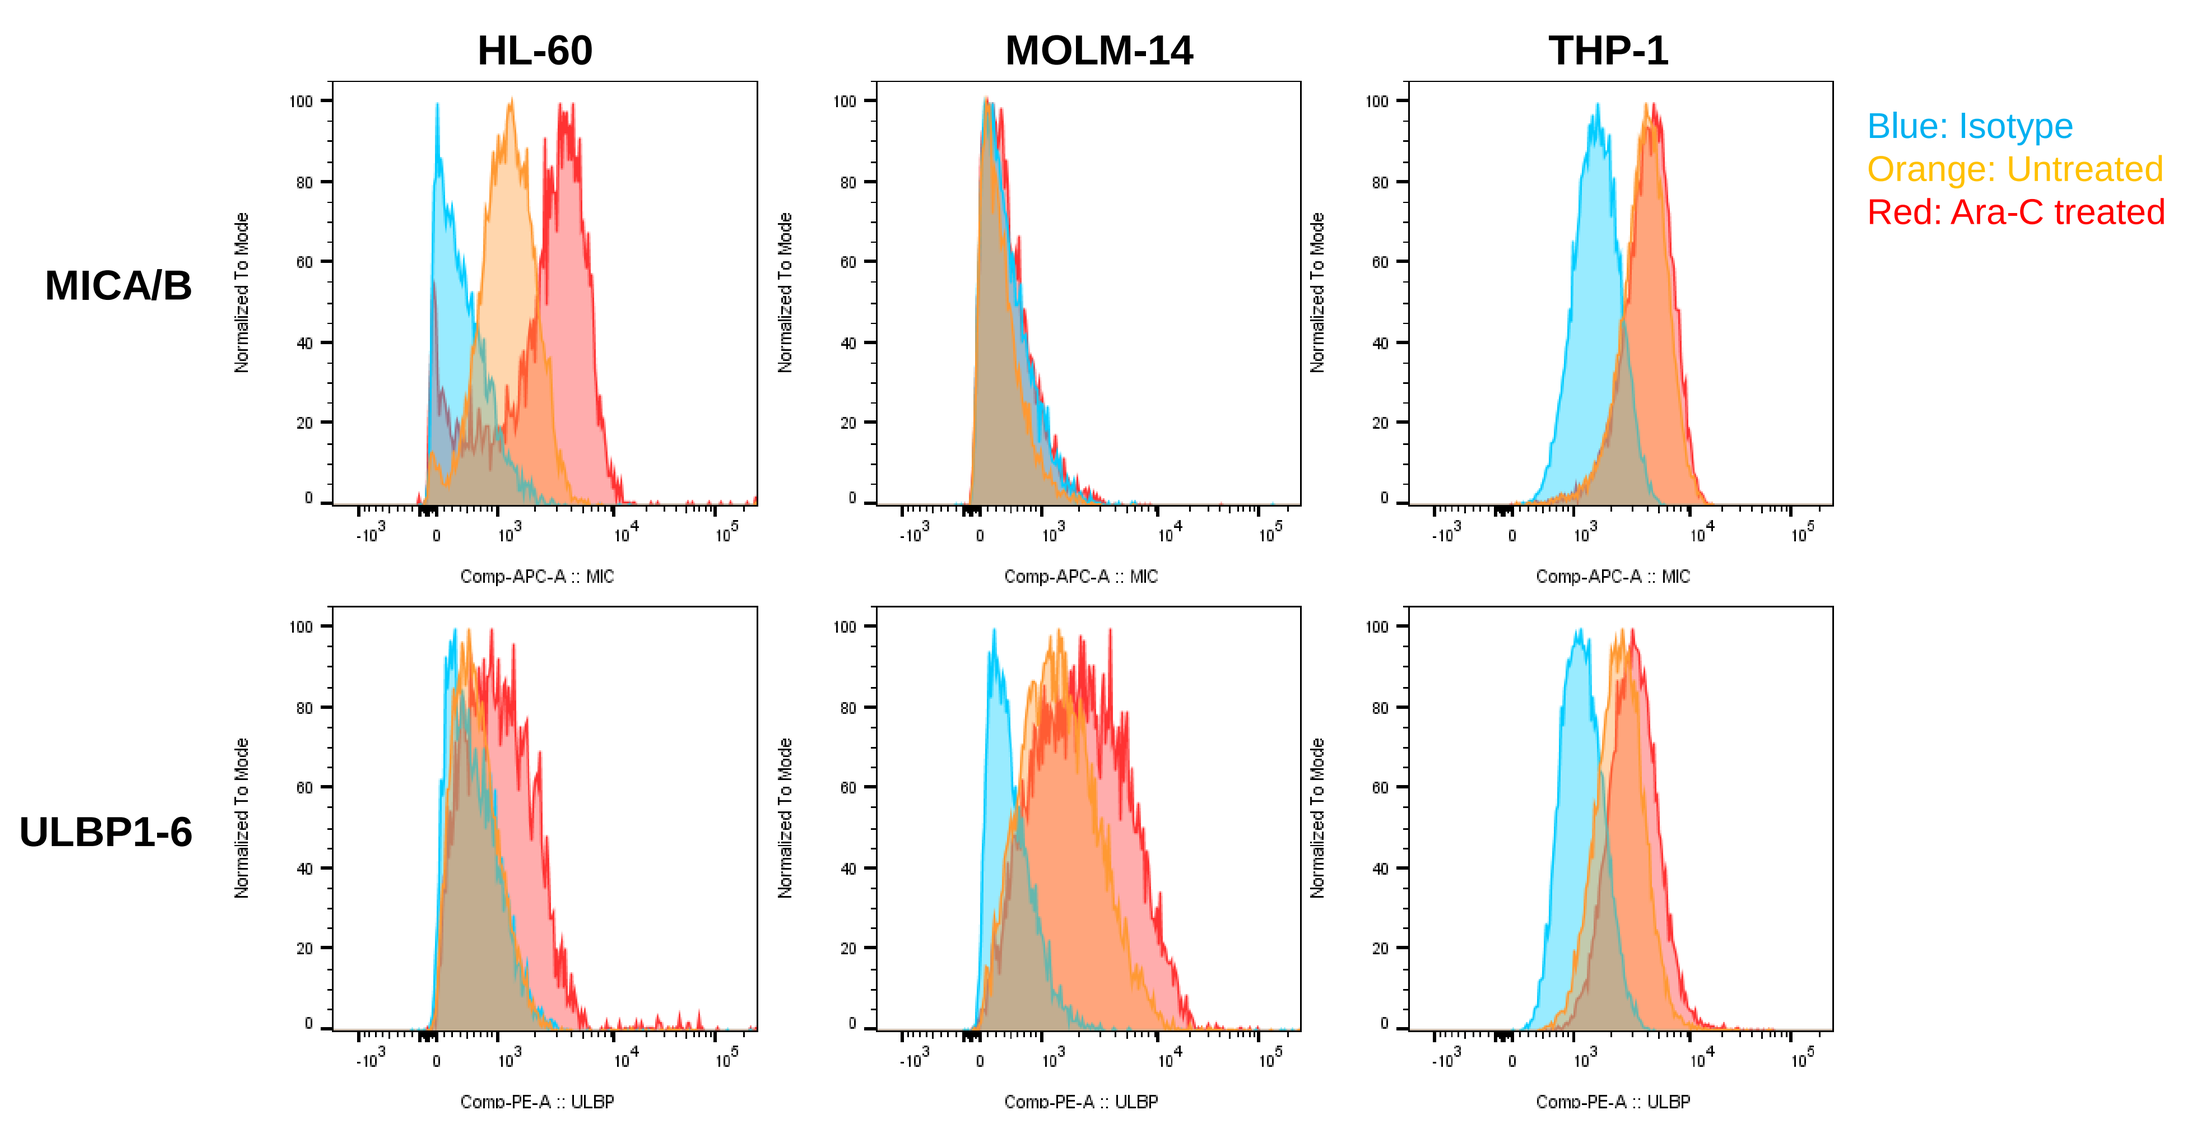
HL-60
MOLM-14
THP-1
MICA/B
ULBP1-6
Blue: Isotype
Orange: Untreated
Red: Ara-C treated

Figure 3. Effect of Ara-C treatment on NKG2D-L expression in AML cell lines. HL-60, MOLM-14, and THP-1 cells were treated with a dose titration of Ara-C for 24 hours. Histograms shown represent the highest level of NKG2D-L expression from the tested dose range.

Ara-C pre-treatment of AML cell lines increases NKX101 anti-leukemic potency

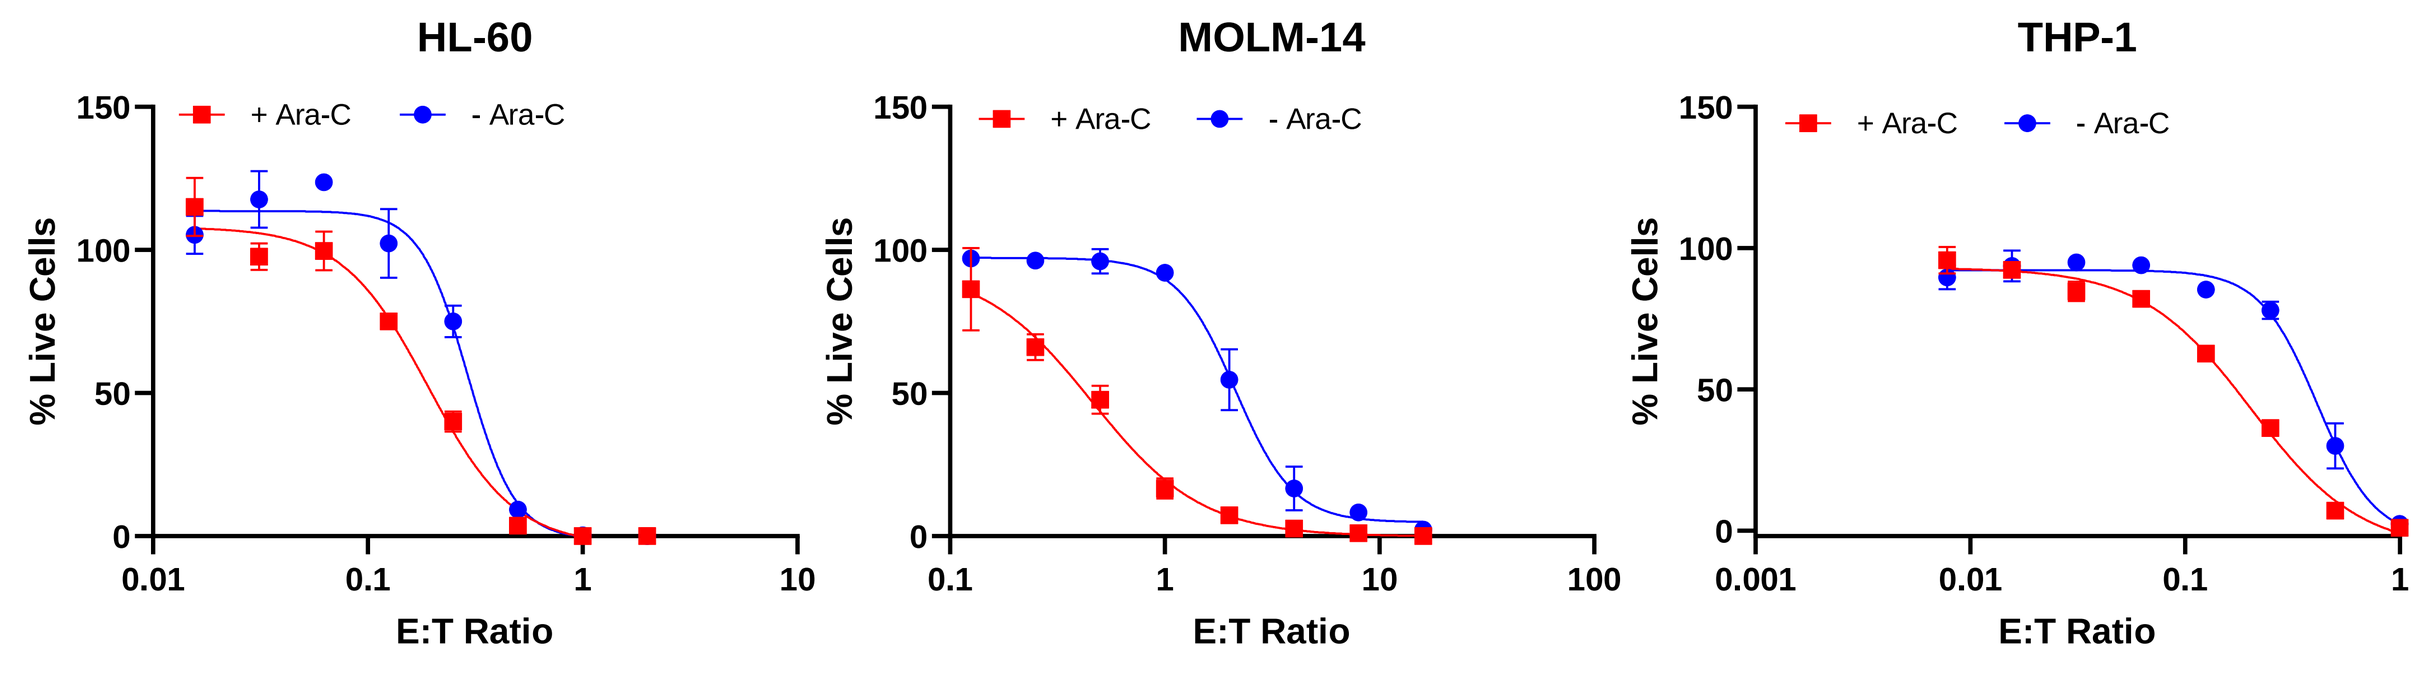
HL-60
MOLM-14
THP-1
+ Ara-C
- Ara-C
% Live Cells
E:T Ratio

AML Cell Line	HL-60	MOLM-14	THP-1
% Increase in Potency over Untreated Cells	53%	366%	103%

Figure 4. Dose-response curves for NKX101 killing of Ara-C pre-treated AML cell lines. HL-60, MOLM-14, and THP-1 were pre-treated with Ara-C for 48 hours at 0.25 uM, 2.15 uM, and 61 uM respectively. Pre-treated target cells were co-cultured with NKX101. Percent increase in potency was calculated using the IC_{50} values from each condition.

Combination of Ara-C and NKX101 demonstrates *in vitro* synergy against AML cell lines

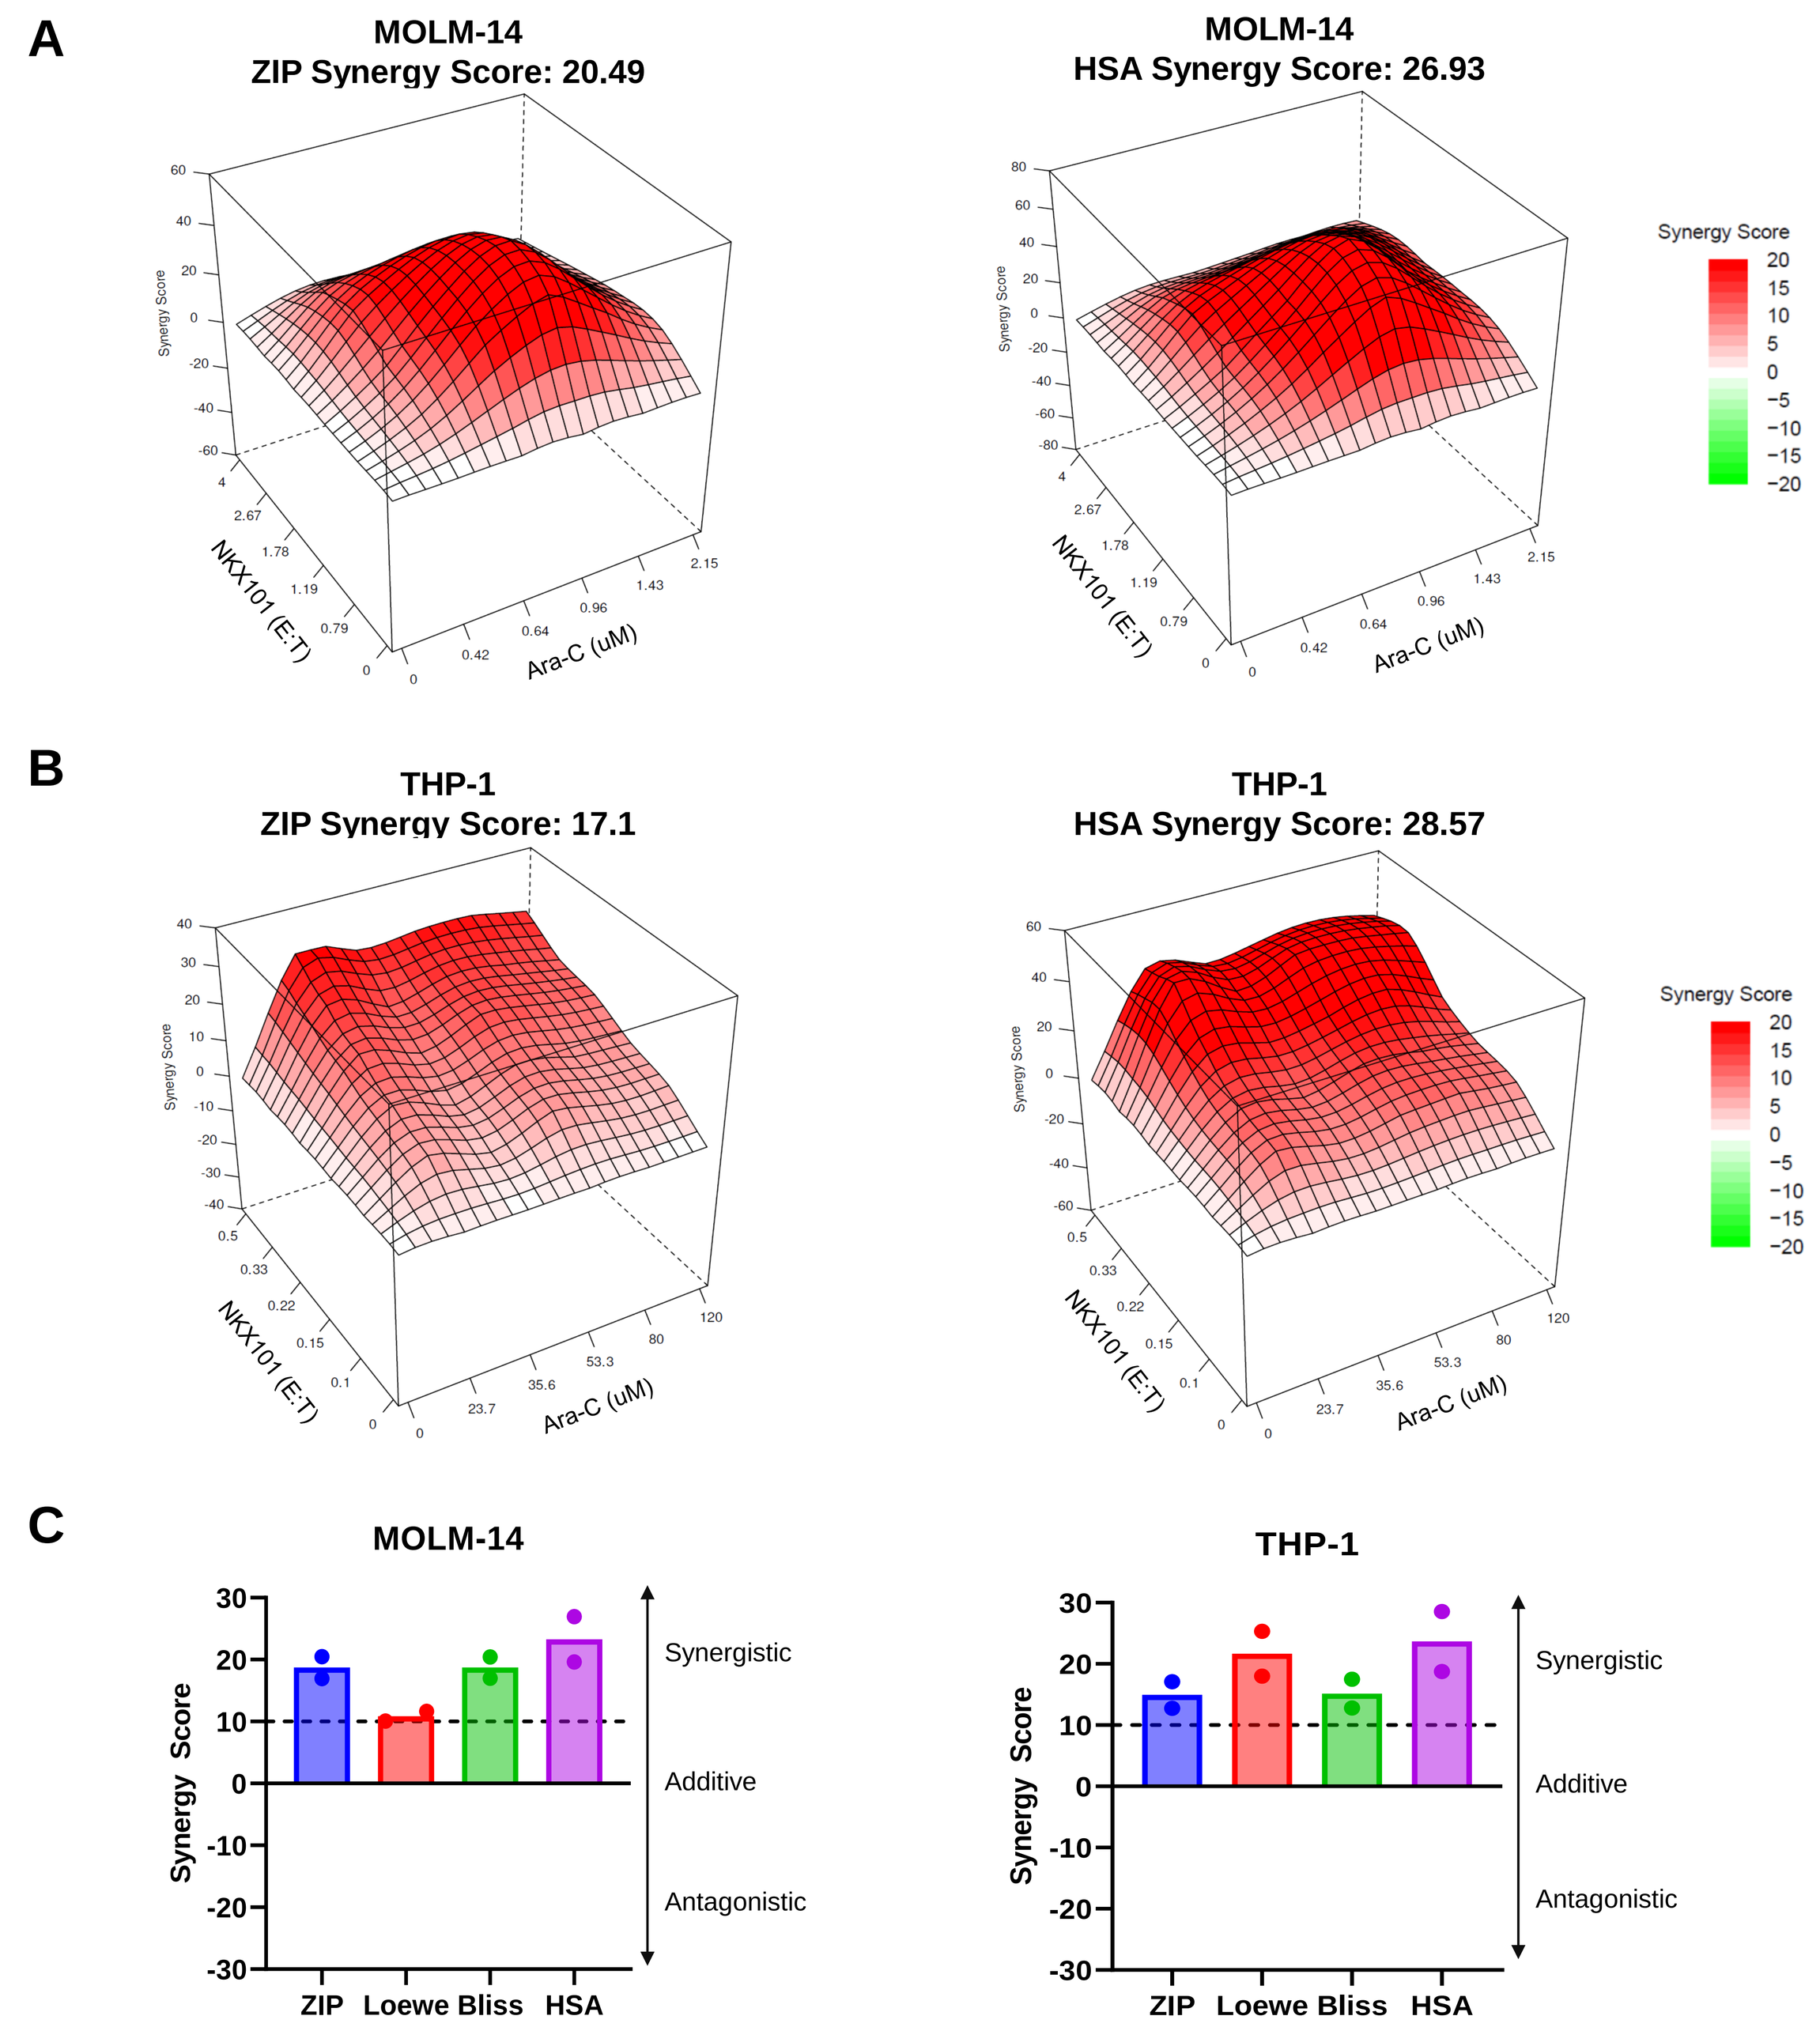
A
MOLM-14
ZIP Synergy Score: 20.49
MOLM-14
HSA Synergy Score: 26.93
B
THP-1
ZIP Synergy Score: 17.1
THP-1
HSA Synergy Score: 28.57
Synergy Score
NKX101 (E:T)
Ara-C (uM)
C
MOLM-14
THP-1
Synergistic
Additive
Antagonistic
ZIP
Loewe
Bliss
HSA

Figure 5. A-B) ZIP and HSA synergy surface maps of a dose-response matrix for the specific killing of MOLM-14 and THP-1 cells by NKX101 following Ara-C treatment. **C)** Summary of ZIP, Loewe, Bliss, and HSA synergy scores from the combination of NKX101 and Ara-C. Data points shown are from two independent experiments.

NKX101 potently kills Ara-C resistant AML cell lines

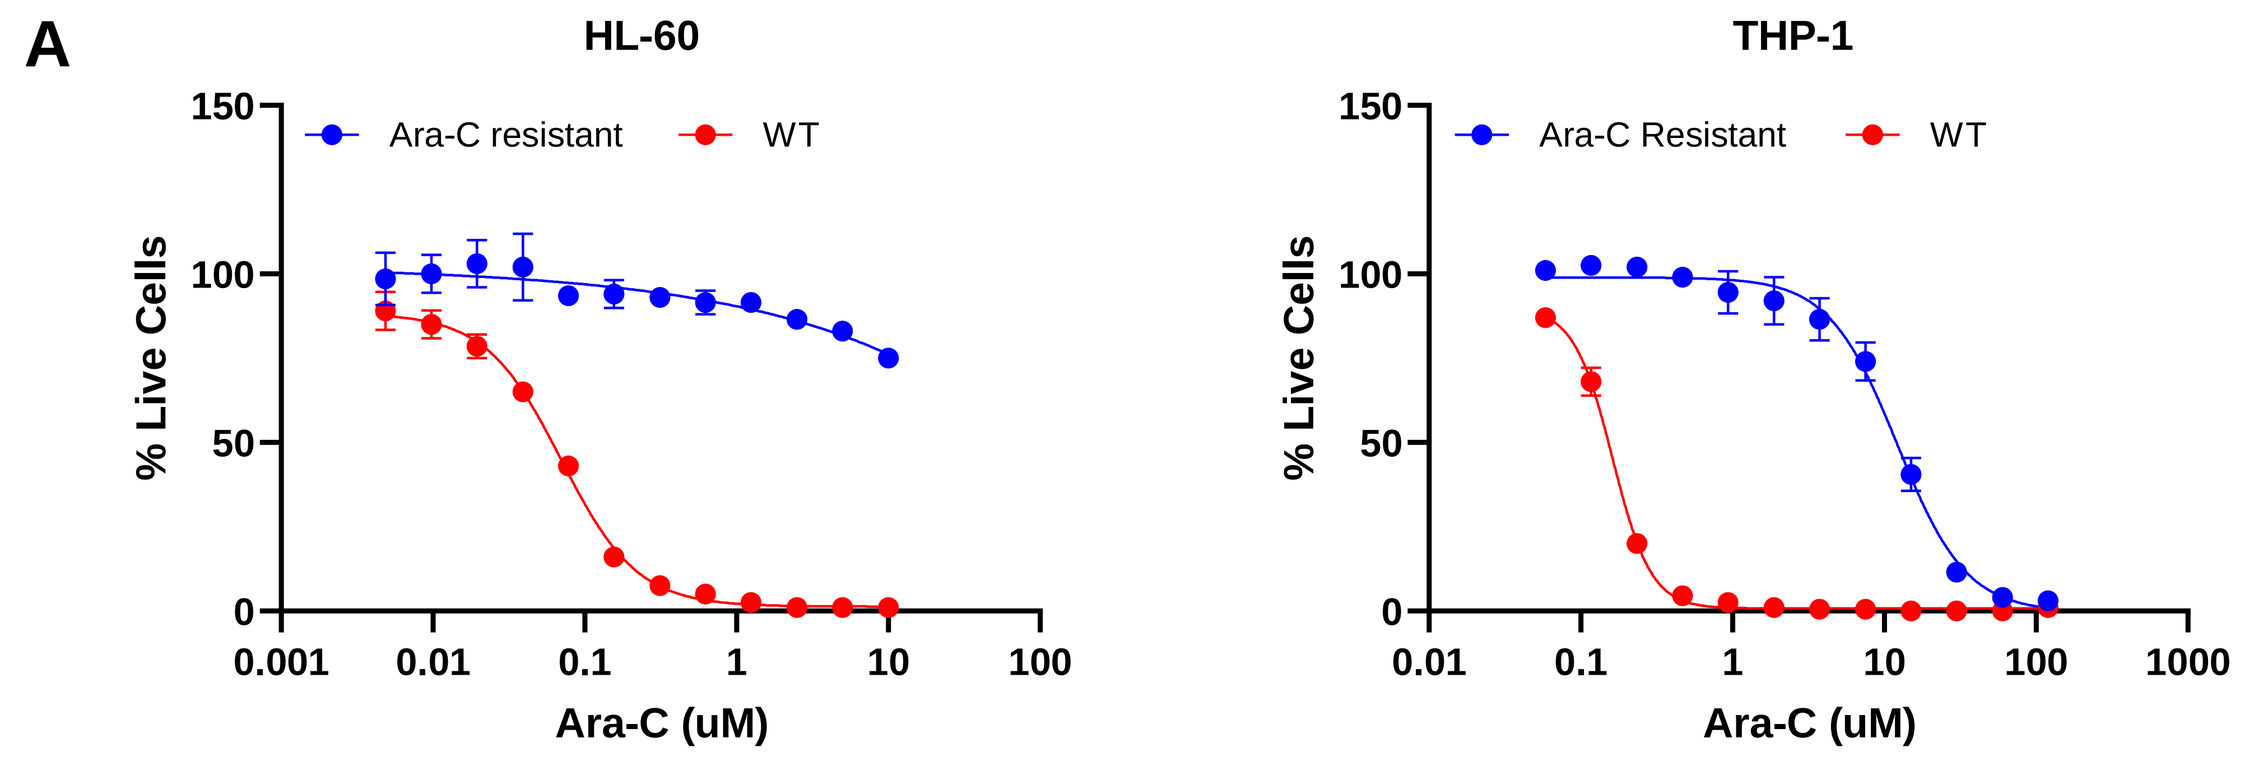
A
HL-60
THP-1
Ara-C resistant
WT
% Live Cells
Ara-C (uM)

	HL-60 Ara-C Resistant	HL-60 Wild Type	THP-1 Ara-C Resistant	THP-1 Wild Type
IC50	Unconverged	0.07	12.2	0.16

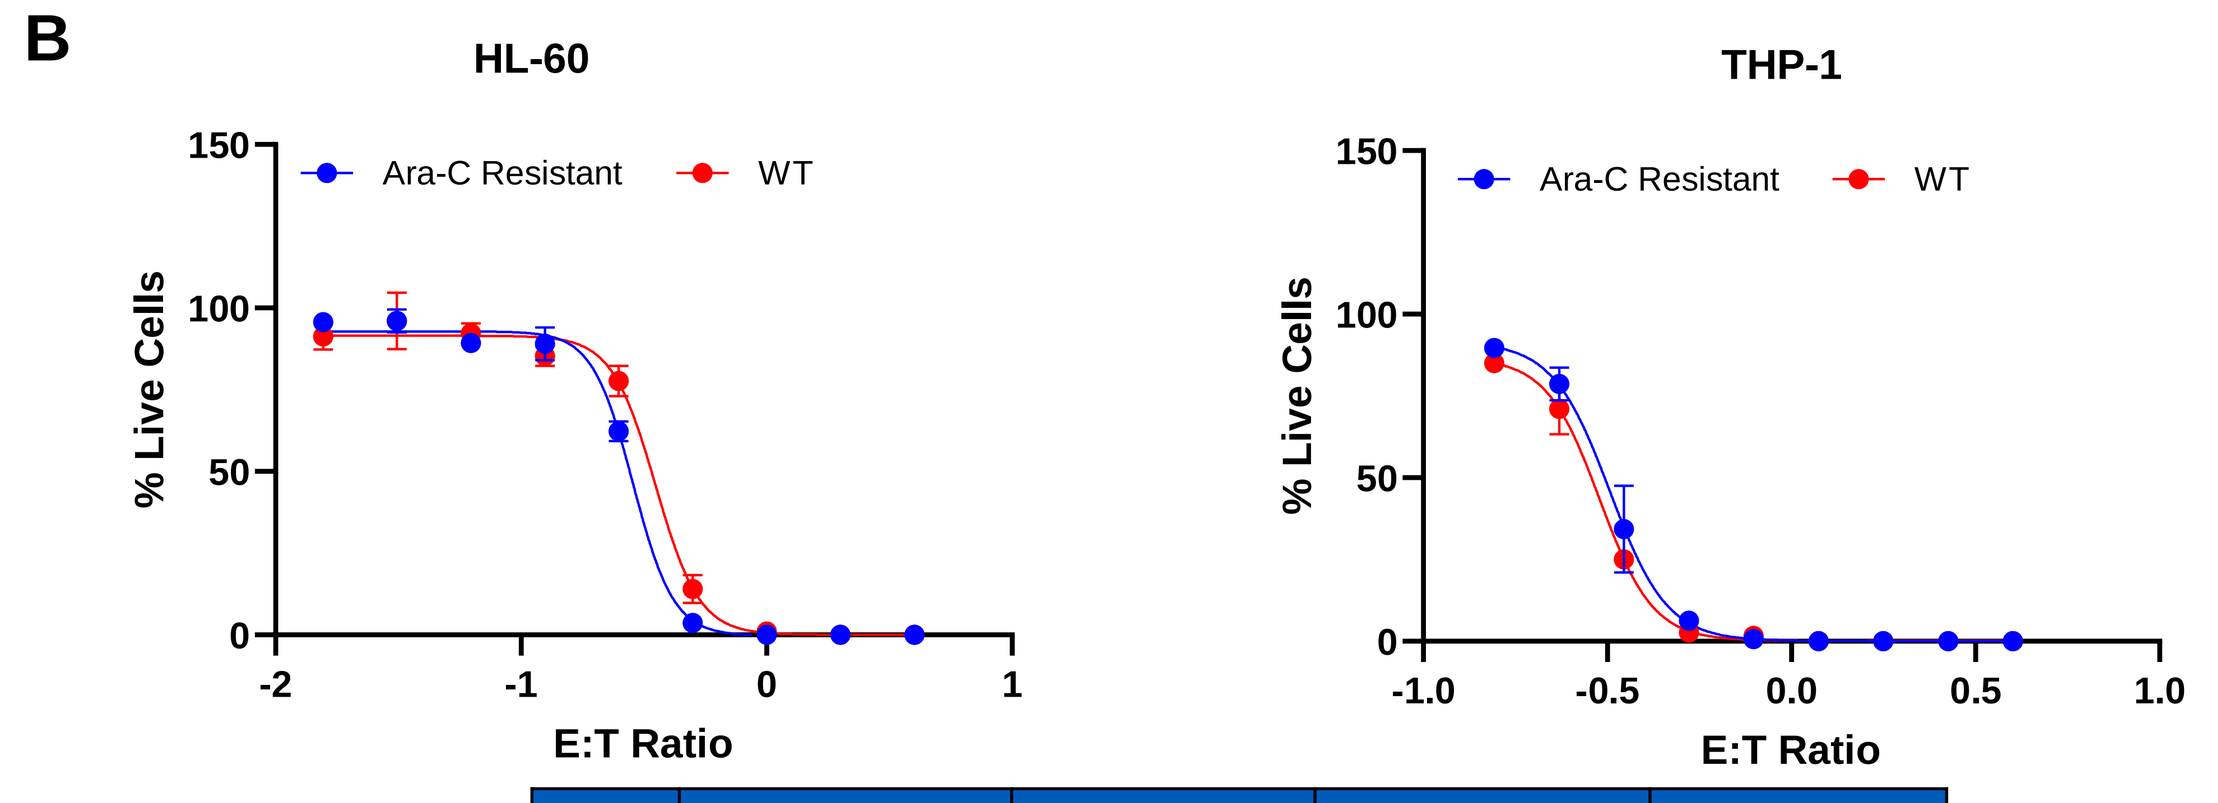
B
HL-60
THP-1
Ara-C Resistant
WT
% Live Cells
E:T Ratio

	HL-60 Ara-C Resistant	HL-60 Wild Type	THP-1 Ara-C Resistant	THP-1 Wild Type
IC50	0.29	0.35	0.32	0.30

Figure 6. A) Treatment of wild type and Ara-C resistant HL-60 and THP-1 cells with increasing concentrations of Ara-C. **B)** Cytotoxicity of NKX101 against wild type and Ara-C resistant HL-60 and THP-1 cells.

Results

To determine if the expression of a single NKG2D ligand is sufficient to trigger NKX101 cytotoxicity, we co-cultured NKX101 with CHO-K1 cells expressing high levels of individual NKG2D-L and observed potent NKX101 cytotoxicity against CHO-K1 cells expressing a single NKG2D-L compared to the CHO-K1 empty vector control cells, producing an approximately 10 to 23-fold enhancement in target cell killing. To evaluate the effects of Ara-C treatment on NKG2D-L expression on AML cell lines, HL-60, MOLM-14, and THP-1 cell lines were cultured with increasing concentrations of Ara-C for 24, 48, and 72 hours. We observed the greatest NKG2D-L upregulation after 24 hours of Ara-C treatment. To evaluate the effect of Ara-C pre-treatment on NKX101 cytotoxic potency, we co-cultured Ara-C pre-treated AML cell lines with a range of NKX101 E:T ratios. NKX101 cells had higher potency against Ara-C pre-treated AML cell lines compared to untreated controls, with a 53%, 103%, and 366% increase in potency observed for HL-60, THP-1 and MOLM-14 cells, respectively. Next, we employed a dose-response matrix to assess synergy scores of NKX101 combined with Ara-C pre-treatment on MOLM-14 and THP-1 cell lines. ZIP and HSA synergy scores ranged from ~17 to ~29, and both Loewe and Bliss synergy scores were above 10. Altogether, the four synergy reference models reveal that NKX101 and Ara-C combine synergistically to kill AML cells in vitro. To evaluate NKX101 activity against Ara-C resistant cell lines, we treated wild type and Ara-C resistant HL-60 and THP-1 cells with a dose-titration of Ara-C and confirmed greater resistance to Ara-C in the resistant cell lines compared to the wild type controls. NKX101 displayed similar potency against both wild type and Ara-C resistant AML cells, demonstrating that cancer cells exhibiting acquired resistance to Ara-C can be eliminated with NKX101 treatment.

Conclusions

In summary, NKX101 efficiently targets and eliminates cells expressing NKG2D-Ls, further validating NKG2D-Ls as therapeutic targets for the treatment of AML and other malignancies. NKX101 cells were potent against NKG2D-L^{+} AML cell lines. Pre-treatment of AML cell lines with Ara-C elevated both their expression of NKG2D-L and their sensitivity to NKX101, demonstrating that the combination can enhance NKX101 cytotoxicity. Utilizing dose-response matrices and the SynergyFinder Plus web application, we determined that the anti-leukemic activity of NKX101 combined with Ara-C is synergistic. Additionally, we observed that Ara-C resistant cell lines can be eliminated by NKX101 with similar potency compared to wild type cells. In summary, we show that combining NKX101 and Ara-C increases the specific killing of AML cells and highlight that NKX101 maintains cytotoxicity against Ara-C resistant cell lines. Altogether, our studies support continued investigation of NKX101 CAR NK cell therapy with modified lymphodepletion incorporating Ara-C for the treatment of r/r AML.

Contact

James Trager, PhD
jtrager@nkartatx.com
www.nkartatx.com